Subset analysis of The HALO Survey evaluating the impact of side effects of medications in patients with glomerular disease

Goldfinch Bio
Saving Kidneys, Ending Dialysis

Grandinetti, A.[1], Campbell, K.[2], Costello, A.[1], Bressler, K.[4], Hammonds, J.[4], Hood, K.[3], Kennedy, J.[1], Melone, L.[1], Mulroy, S.[4], Watson, D.[4], Duhaime, E[1].

[1]Goldfinch Bio, Cambridge, MA, United States. [2] Icahn School of Medicine at Mount Sinai, New York, NY, United States. [3] NephCure Kidney International, King of Prussia, PA, United States.
[4] Patient Advisory Board, Goldfinch Bio, MA, United States.

1. INTRODUCTION

Despite the dramatic rise in and prevalence of CKD in recent years, including glomerular diseases (GD), there are virtually no therapies approved to treat the underlying causes of most GD. Several approved therapies are used off-label but only treat disease symptoms, such as high blood pressure and protein loss (angiotensin-converting enzyme, or ACE, inhibitors and angiotensin II receptor blockers, or ARBs), swelling (diuretics), and overactivation of the immune system (corticosteroids, calcineurin inhibitors). These drugs are not disease-modifying, and their mechanisms work through systemic versus targeted actions, often resulting in systemic side effects and downstream complications. Common side effects include fatigue, insomnia, weight gain, acne, mood swings, osteoporosis, cataracts, and tremors, among others.

Goldfinch Bio (GFB) developed the "Assessment of **H**ealth and qu**A**lity of **L**ife impacts of standard treatments **O**n patients with glomerular diseases and their caregivers" (HALO Survey), based on insights from patient focus groups and the GFB Patient Advisory Board (PAB), to explore the impact of side effects (SE) of GD treatments as reported by patients with GD and their caregivers. While research highlighting the quality of life (QOL) impact current standard medications have on this patient community is very limited, available evidence suggests that current treatments for GD and associated SE have a negative impact on patients' QOL. We herein report the findings of a subset of The HALO survey respondents (n=62) who were pre-end-stage kidney disease (ESKD).

2. METHODS

The HALO Survey was developed and reviewed in partnership with the PAB, NephCure, and a U.S.-based academic nephrologist to ensure accurate representation from all stakeholders. People diagnosed with focal segmental glomerulosclerosis (FSGS), IgA nephropathy (IGAN), membranous nephropathy (MN), or minimal change disease (MCD) ≥18 years old, or caregivers of pediatric patients with these diseases (<18) were eligible to participate in the survey. Questions were developed to avoid negative response bias. Survey software was licensed (Qualtrics, Provo, UT) to administer The HALO Survey. The 62 pre-ESKD patients were selected for analysis as they represented the most homogenous subset within the survey population. Of note, for purposes of this analysis, "patients" refers to both patient and caregiver survey respondents.

.

Example Patient Quotes Summarizing Overall Experiences and Biggest Impact to Life with Medications
"The worst part was feeling suicidal for months on end from ultra-high dose steroids (I did the Tune-Mendoza protocol twice)."
"Apart from ACE inhibitors, the medications make me feel worse than the actual disease."
"Taking these drugs for a long time has changed my face and hair. I became old and have lack of vitality, which hinders many of my social activities. I have become not confident and lonely."
"I feel like there really are no medications that treat my FSGS and that I am forced to take medicine that destroys my body to treat it."
"I understand the need for medication to prevent further kidney damage but as of right now the side effects are outweighing the benefits."
"Drugs have been necessary but in the last 20 years had little sustainable impact on FSGS as I've lost two kidney transplants. I now have nocturnal dialysis every other night and am not considering another transplant until there is a cure for FSGS."
"The choices are very harmful; the tradeoff is frustrating… savings kidneys to what end?"
One patient described medications as "incredibly frustrating and despair-provoking to destroy the rest of my physical and mental well-being through treatments that didn't even work."

3. RESULTS

The survey was open from 10/5/21 to 11/1/21. Of the 392 responses received, 257 had ESKD, 62 were pre-ESKD (pre-dialysis and pre-transplant), and 73 were incomplete or deemed ineligible. The following results refer to the 62 pre-ESKD subset. All respondents took at least 1-2 medications, while 34% took ≥3. Patients were asked to rate their overall experience of medications on a 7-point scale, from extremely positive to extremely negative. 76% of all patients said their overall experiences were extremely negative to neutral. The most commonly reported SE/concerns that negatively impacted patients were mood swings, fear of getting sick/immunosuppression, hair loss, and brain fog. 56% indicated that medications affected their mental health; 40% reported having ≥2 mental health issues. 15% of patients reported that they missed 15-30 days of school or work in the past 12 months due to kidney disease. 18% of patients reported that they missed 31 or more days of school or work in the same timeframe. 27% of adult patients (n=17) indicated that they currently do not work due to kidney disease.

Disease-Specific Analysis

We conducted further analysis of the 62 pre-ESKD patient subset based on specific disease (29 FSGS, 16 IGAN, 0 MN, and 17 MCD patients). FSGS patients reported the most negative overall experience using medications. 100% of FSGS patients described their overall experience of medications as extremely negative to neutral, compared to IGAN (44%) and MCD (65%) patients. 29% of FSGS patients reported that medications always bothered them, compared to 0% of IGAN and 6% of MCD patients. 59% of adult (>18) FSGS patients (n=22) reported not currently having a job due to kidney disease compared to 25% of IGAN (n=14) and 18% of MCD (n=15) adult patients.

4. CONCLUSION

The findings from this subset analysis of The HALO Survey suggest that current standard treatments for GD contribute to poor QOL. Most GD patients reported negative experiences with their medications and were frequently or always bothered by SE. This analysis revealed that medication SE led to intense physical and mental burdens on patients. Patients also reported that the medications had a significant limiting impact on their productivity, including a significant amount of missed work/school as well as missed important life events. Patients with FSGS had the most negative overall experience using medications compared to patients with other GD.

Given the significant underserved medical needs of GD patients and poor QOL that current medication side effects have, novel approaches and treatments are needed that can prevent patients' disease progression to ESKD accompanied by improved QOL. Future drug development should focus on targeted therapies that improve kidney health while minimizing systemic SE to enhance QOL.

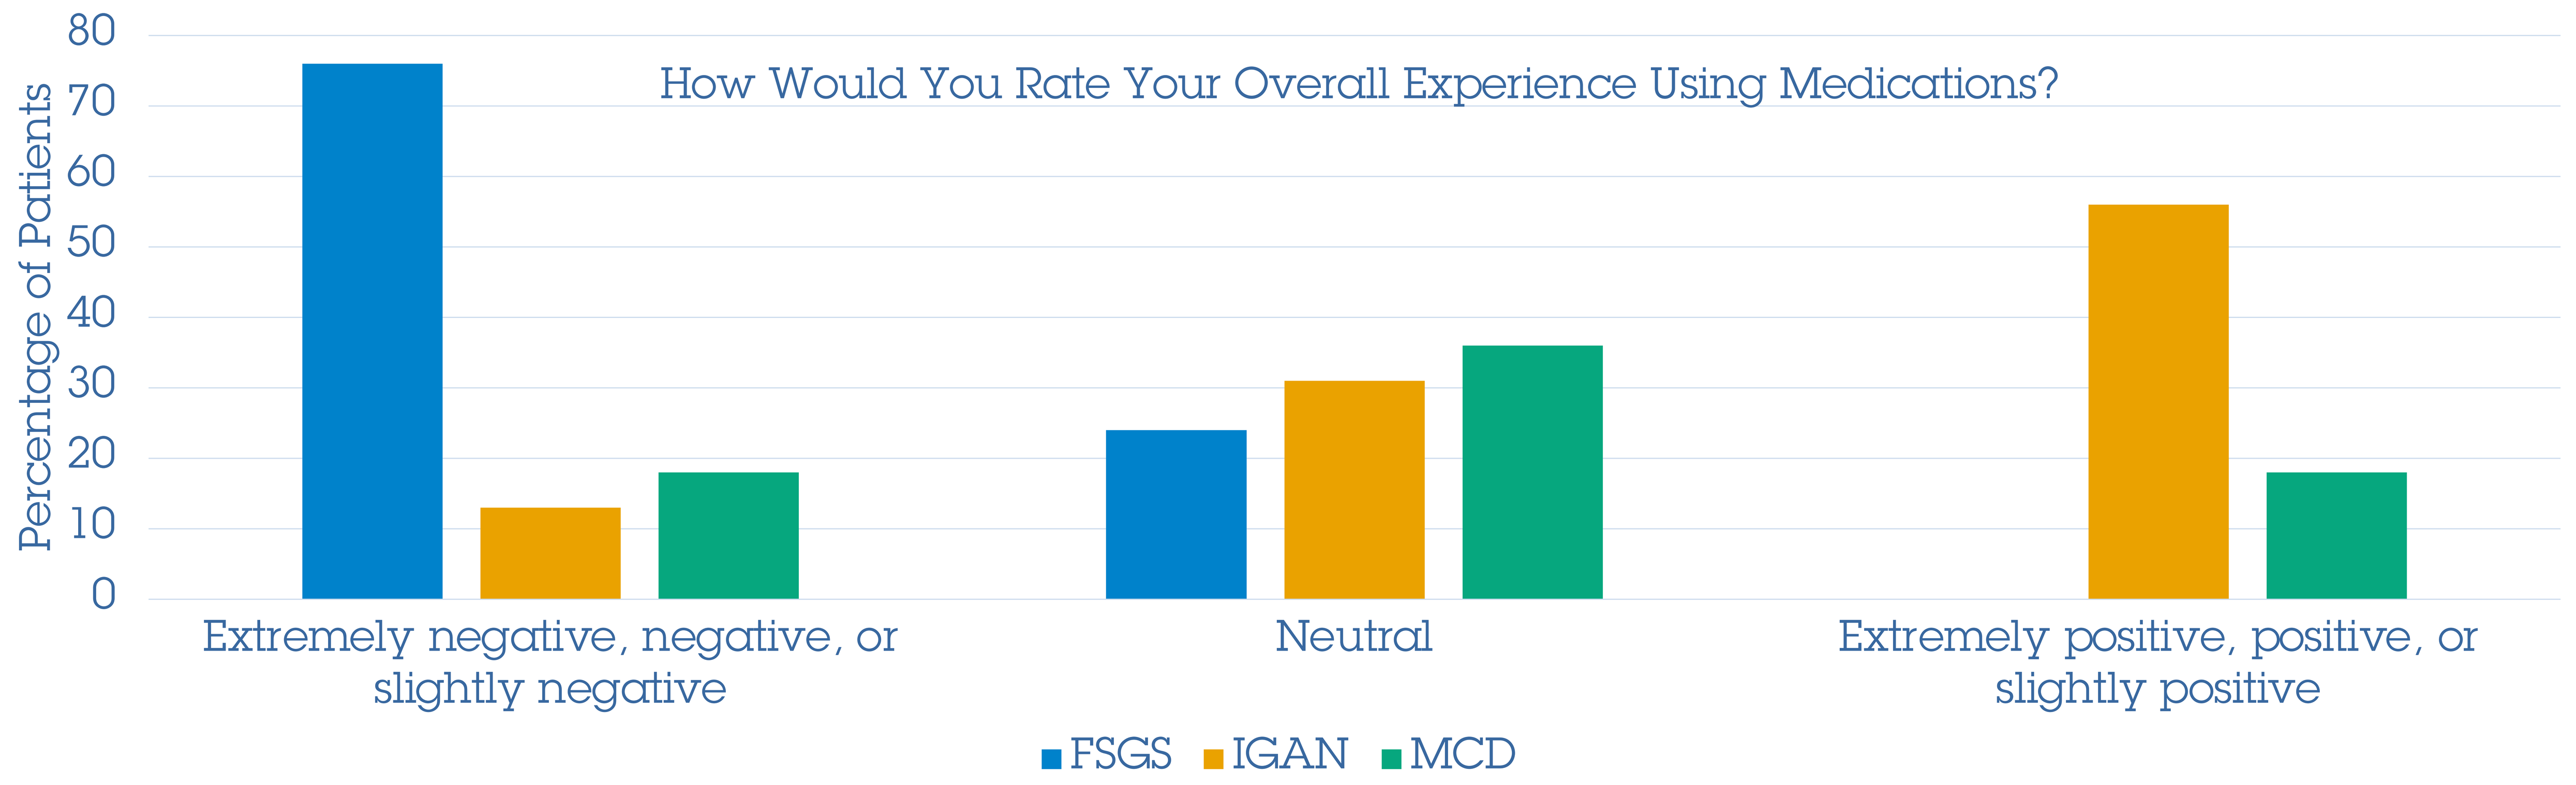
How Would You Rate Your Overall Experience Using Medications?
Percentage of Patients
0
10
20
30
40
50
60
70
80
Extremely negative, negative, or slightly negative
Neutral
Extremely positive, positive, or slightly positive
FSGS
IGAN
MCD

Presented at National Kidney Foundation Spring Clinical Meeting | 6-10 April 2022

Contact: agrandinetti@goldfinchbio.com